Risk and protective factors for cognitive performance and decline in diverse ethno-regional groups: the COSMIC collaboration

Darren Lipnicki[1], Steve Makkar[1], John Crawford[1], Anbupalam Thalamuthu[1], Nicole Kochan[1], Henry Brodaty[1,2], Perminder Sachdev[1,2], for Cohort Studies of Memory in an International Consortium (COSMIC)

[1] Centre for Healthy Brain Ageing (CHeBA), School of Psychiatry, Faculty of Medicine, University of New South Wales, Australia; [2] Primary Dementia Collaborative Research Centre, School of Psychiatry, Faculty of Medicine, University of New South Wales, Australia.

Objective

Cognitive decline, whether a feature of normative ageing or dementia, imposes enormous financial costs and societal burden globally. With no effective treatments, evidence for risk and protective factors is paramount, with view to potential prevention. We investigated a comprehensive set of such factors on a truly international scale, using longitudinal cohort data shared by members of the COSMIC (Cohort Studies of Memory in an International Consortium) collaboration.

Method

- Data were from 20 cohorts across 15 countries and 5 continents (Table 1), for 48,522 individuals aged 54–105 years (58.4% female) and non-demented at baseline. Each study had 2–16 assessment waves (median = 3) and a follow-up duration of 2–15 years.
- Primary outcome measures were standardized scores for the Mini-Mental State Examination (MMSE) and global cognition (averaged across tests of memory, language, attention/processing speed, and executive functioning).
- The harmonized factors investigated were age, sex, education, alcohol consumption (nil, 1 drink/week, 2+ drinks/week), anxiety, apolipoprotein ε4 allele (*APOE*4*) status, atrial fibrillation, blood and pulse pressure, body mass index (BMI), cardiovascular disease (other than atrial fibrillation), depression (both current and history of), diabetes, health (self-reported: very good, good, poor), high cholesterol, hypertension, peripheral vascular disease, physical activity (minimal, moderate at least once a week, vigorous at least once a week), smoking (never, past, current), and stroke history.
- Single factor and combined factor linear mixed models were followed by IPD meta-analyses that pooled effects across cohorts.
- We also compared associations for white and Asian groups, based on all whites from 11 predominantly white cohorts (N = 25,174) and all Asians from 5 cohorts (N = 10,296). Individuals from the Latin American and Sacramento cohorts were not included.

Table 1. Contributing study name, abbreviation, and country

Study	Abbreviation	Location
Bambui Cohort Study of Aging	Bambui	Brazil
Cognitive Function & Ageing Study	CFAS	United Kingdom
Cuban Health and Alzheimer Study	CHAS	Cuba
Einstein Aging Study	EAS	USA
Etude Santé Psychologique et Traitement	ESPRIT	France
Hellenic Longitudinal Investigation of Aging and Diet	HELIAD	Greece
Hong Kong Memory and Ageing Prospective Study	HK-MAPS	Hong Kong
Invecchiamento Cerebrale in Abbiategrasso	Invece.Ab	Italy
Korean Longitudinal Study on Cognitive Aging and Dementia	KLOSCAD	South Korea
Leipzig Longitudinal Study of the Aged	LEILA75+	Germany
Maastricht Ageing Study	MAAS	The Netherlands
Monongahela Valley Independent Elders Survey	MoVIES	USA
Personality and Total Health Through Life Project	PATH	Australia
Sacramento Area Latino Study on Aging	SALSA	USA
São Paulo Ageing & Health Study	SPAH	Brazil
Sasaguri Genkimon Study	SGS	Japan
Singapore Longitudinal Ageing Studies (I)	SLASI	Singapore
Sydney Memory and Ageing Study	Sydney MAS	Australia
Tajiri Project	Tajiri	Japan
Zaragoza Dementia Depression Project	ZARADEMP	Spain

Results

Cognitive performance and decline (Table 2.)

Table 2. Factors showing significant associations with cognitive performance or decline[a]

Cognitive performance						
	Single factor models		Combined model 1[b]		Combined model 2[c]	
Factor	Global cognition	MMSE	Global cognition	MMSE	Global cognition	MMSE
Age at baseline	-0.102 (0.035)**	-0.058 (0.028)*	-0.091 (0.012)***	-0.053 (0.007)***	-0.103 (0.013)***	-0.060 (0.009)***
Education	0.138 (0.013)***	0.092 (0.013)***	0.115 (0.021)***	0.095 (0.011)***	0.115 (0.021)***	0.095 (0.011)***
Alcohol, 1 drink/week[d]	0.118 (0.083)	0.183 (0.073)*	0.077 (0.108)	0.161 (0.085)	0.080 (0.113)	0.148 (0.087)
Alcohol, 2+ drinks/week[d]	0.072 (0.047)	0.126 (0.055)*	0.081 (0.110)	0.069 (0.051)	0.064 (0.111)	0.063 (0.049)
Anxiety	-0.15 (0.073)*	-0.105 (0.055)				
*APOE*4* carrier	-0.197 (0.062)**	-0.037 (0.034)	-0.311 (0.107)**	-0.006 (0.055)	-0.251 (0.101)*	-0.078 (0.049)
Body mass index[e]	-0.002 (0.001)*	-0.001 (0.0004)			0.001 (0.007)	-0.002 (0.006)
Cardiovascular disease	-0.050 (0.071)	0.030 (0.030)	0.030 (0.058)	0.095 (0.044)*	0.027 (0.062)	0.141 (0.047)**
Depression, current	-0.275 (0.099)**	-0.146 (0.037)***			-0.113 (0.057)*	-0.073 (0.069)
Diabetes	-0.294 (0.052)***	-0.152 (0.028)***	-0.230 (0.097)*	-0.119 (0.085)	-0.209 (0.116)	-0.136 (0.084)
Health, poor[f]	-0.558 (0.097)***	-0.255 (0.065)***				
Hypertension	-0.109 (0.05)*	-0.020 (0.019)	-0.170 (0.115)	-0.007 (0.044)	-0.206 (0.122)	-0.022 (0.044)
Peripheral vascular disease	-0.298 (0.131)*	-0.044 (0.052)				
Physical activity, vigorous[g]	0.211 (0.066)**	0.205 (0.051)***			0.160 (0.146)	0.168 (0.061)**
Smoking, current[h]	-0.180 (0.125)	-0.119 (0.056)	0.081 (0.110)	0.069 (0.051)	-0.134 (0.167)	-0.202 (0.082)*
Stroke history	-0.344 (0.113)**	-0.337 (0.058)***	-0.252 (0.249)	-0.223 (0.089)*	-0.218 (0.270)	-0.049 (0.041)
Cognitive decline						
	Single factor models		Combined model 1[b]		Combined model 2[c]	
Factor	Global cognition	MMSE	Global cognition	MMSE	Global cognition	MMSE
Age at baseline	-0.007 (0.001)***	-0.005 (0.001)***	-0.007 (0.001)***	-0.005 (0.001)***	-0.007 (0.006)***	-0.005 (0.005)***
Alcohol, 2+ drinks/week[d]	0.011 (0.007)	0.012 (0.005)**	0.012 (0.014)	0.001 (0.008)	0.005 (0.016)	0.000 (0.009)
Anxiety	0.028 (0.017)	0.011 (0.007)				
*APOE*4* positive	-0.028 (0.008)***	-0.009 (0.005)	-0.034 (0.014)*	-0.010 (0.008)	-0.041 (0.018)*	-0.017 (0.008)*
Cardiovascular disease	0.008 (0.006)	0.010 (0.004)**	0.014 (0.008)	0.016 (0.007)*	0.017 (0.008)*	0.018 (0.007)**
Diabetes	-0.013 (0.011)	-0.008 (0.007)	-0.001 (0.010)	-0.013 (0.013)	-0.001 (0.011)	-0.019 (0.009)*
Health, poor[f]	-0.024 (0.011)*	-0.004 (0.006)				
Smoking, past[h]	-0.010 (0.005)*	0.000 (0.003)	-0.008 (0.022)	-0.009 (0.018)	-0.007 (0.009)	-0.001 (0.009)
Stroke history	-0.038 (0.018)*	-0.023 (0.010)*	-0.012 (0.007)	-0.001 (0.007)	-0.024 (0.022)	-0.028 (0.015)
Baseline score	0.004 (0.003)	-0.004 (0.002)*	-0.003 (0.005)	0.001 (0.005)	0.002 (0.004)	0.001 (0.004)

[a] Results are B (SE) at the mean time in study (3.1 y) and controlled for age at baseline (mean = 72.7 y), education (mean = 9.0 y) and sex (42% male); -B indicates worse performance or more decline. *p < 0.05, **p < 0.01, ***p < 0.001.
[b] Used data from 11 cohorts (n = 13917) and included alcohol consumption, *APOE*4* status, cardiovascular disease, diabetes, high cholesterol, hypertension, smoking, and stroke history. The model was repeated with *APOE*4* status replaced with either BMI or current depression (n = 17270 and 18011, respectively).
[c] Used data from nine cohorts (n = 11897) and included alcohol consumption, *APOE*4* status, BMI, cardiovascular disease, depression, diabetes, high cholesterol, hypertension, physical activity, smoking, and stroke history.
[d] vs. nil/minimal alcohol.
[e] Centred at mean = 25.2 kg/m^2.
[f] vs. very good.
[g] 1+ times/week vs. minimal activity.
[h] vs. never smoked.

Changing rates of decline over time in study (significant quadratic effects): Accelerating decline was found for *APOE*4* carriage, and for individuals with good health (vs very good health). Conversely, slowing rates of decline were found for high cholesterol, diabetes, peripheral vascular disease, good and poor health, BMI, and current smoking.

White vs Asian differences

Cognitive performance (Fig 1): Global cognition scores for males were lower than females among whites but higher than females among Asians. For the MMSE, *APOE*4* carriers had higher scores than non-carriers among Asians but not among whites, and poor health was associated with lower scores among whites but not among Asians. Compared to whites, Asians showed greater negative effects on MMSE scores associated with ever smoking.
Cognitive decline (Fig 2): Compared to whites, Asians showed more decline in MMSE scores associated with diabetes, as well as a growing rate of decline associated with each of diabetes, high cholesterol and increasing education.

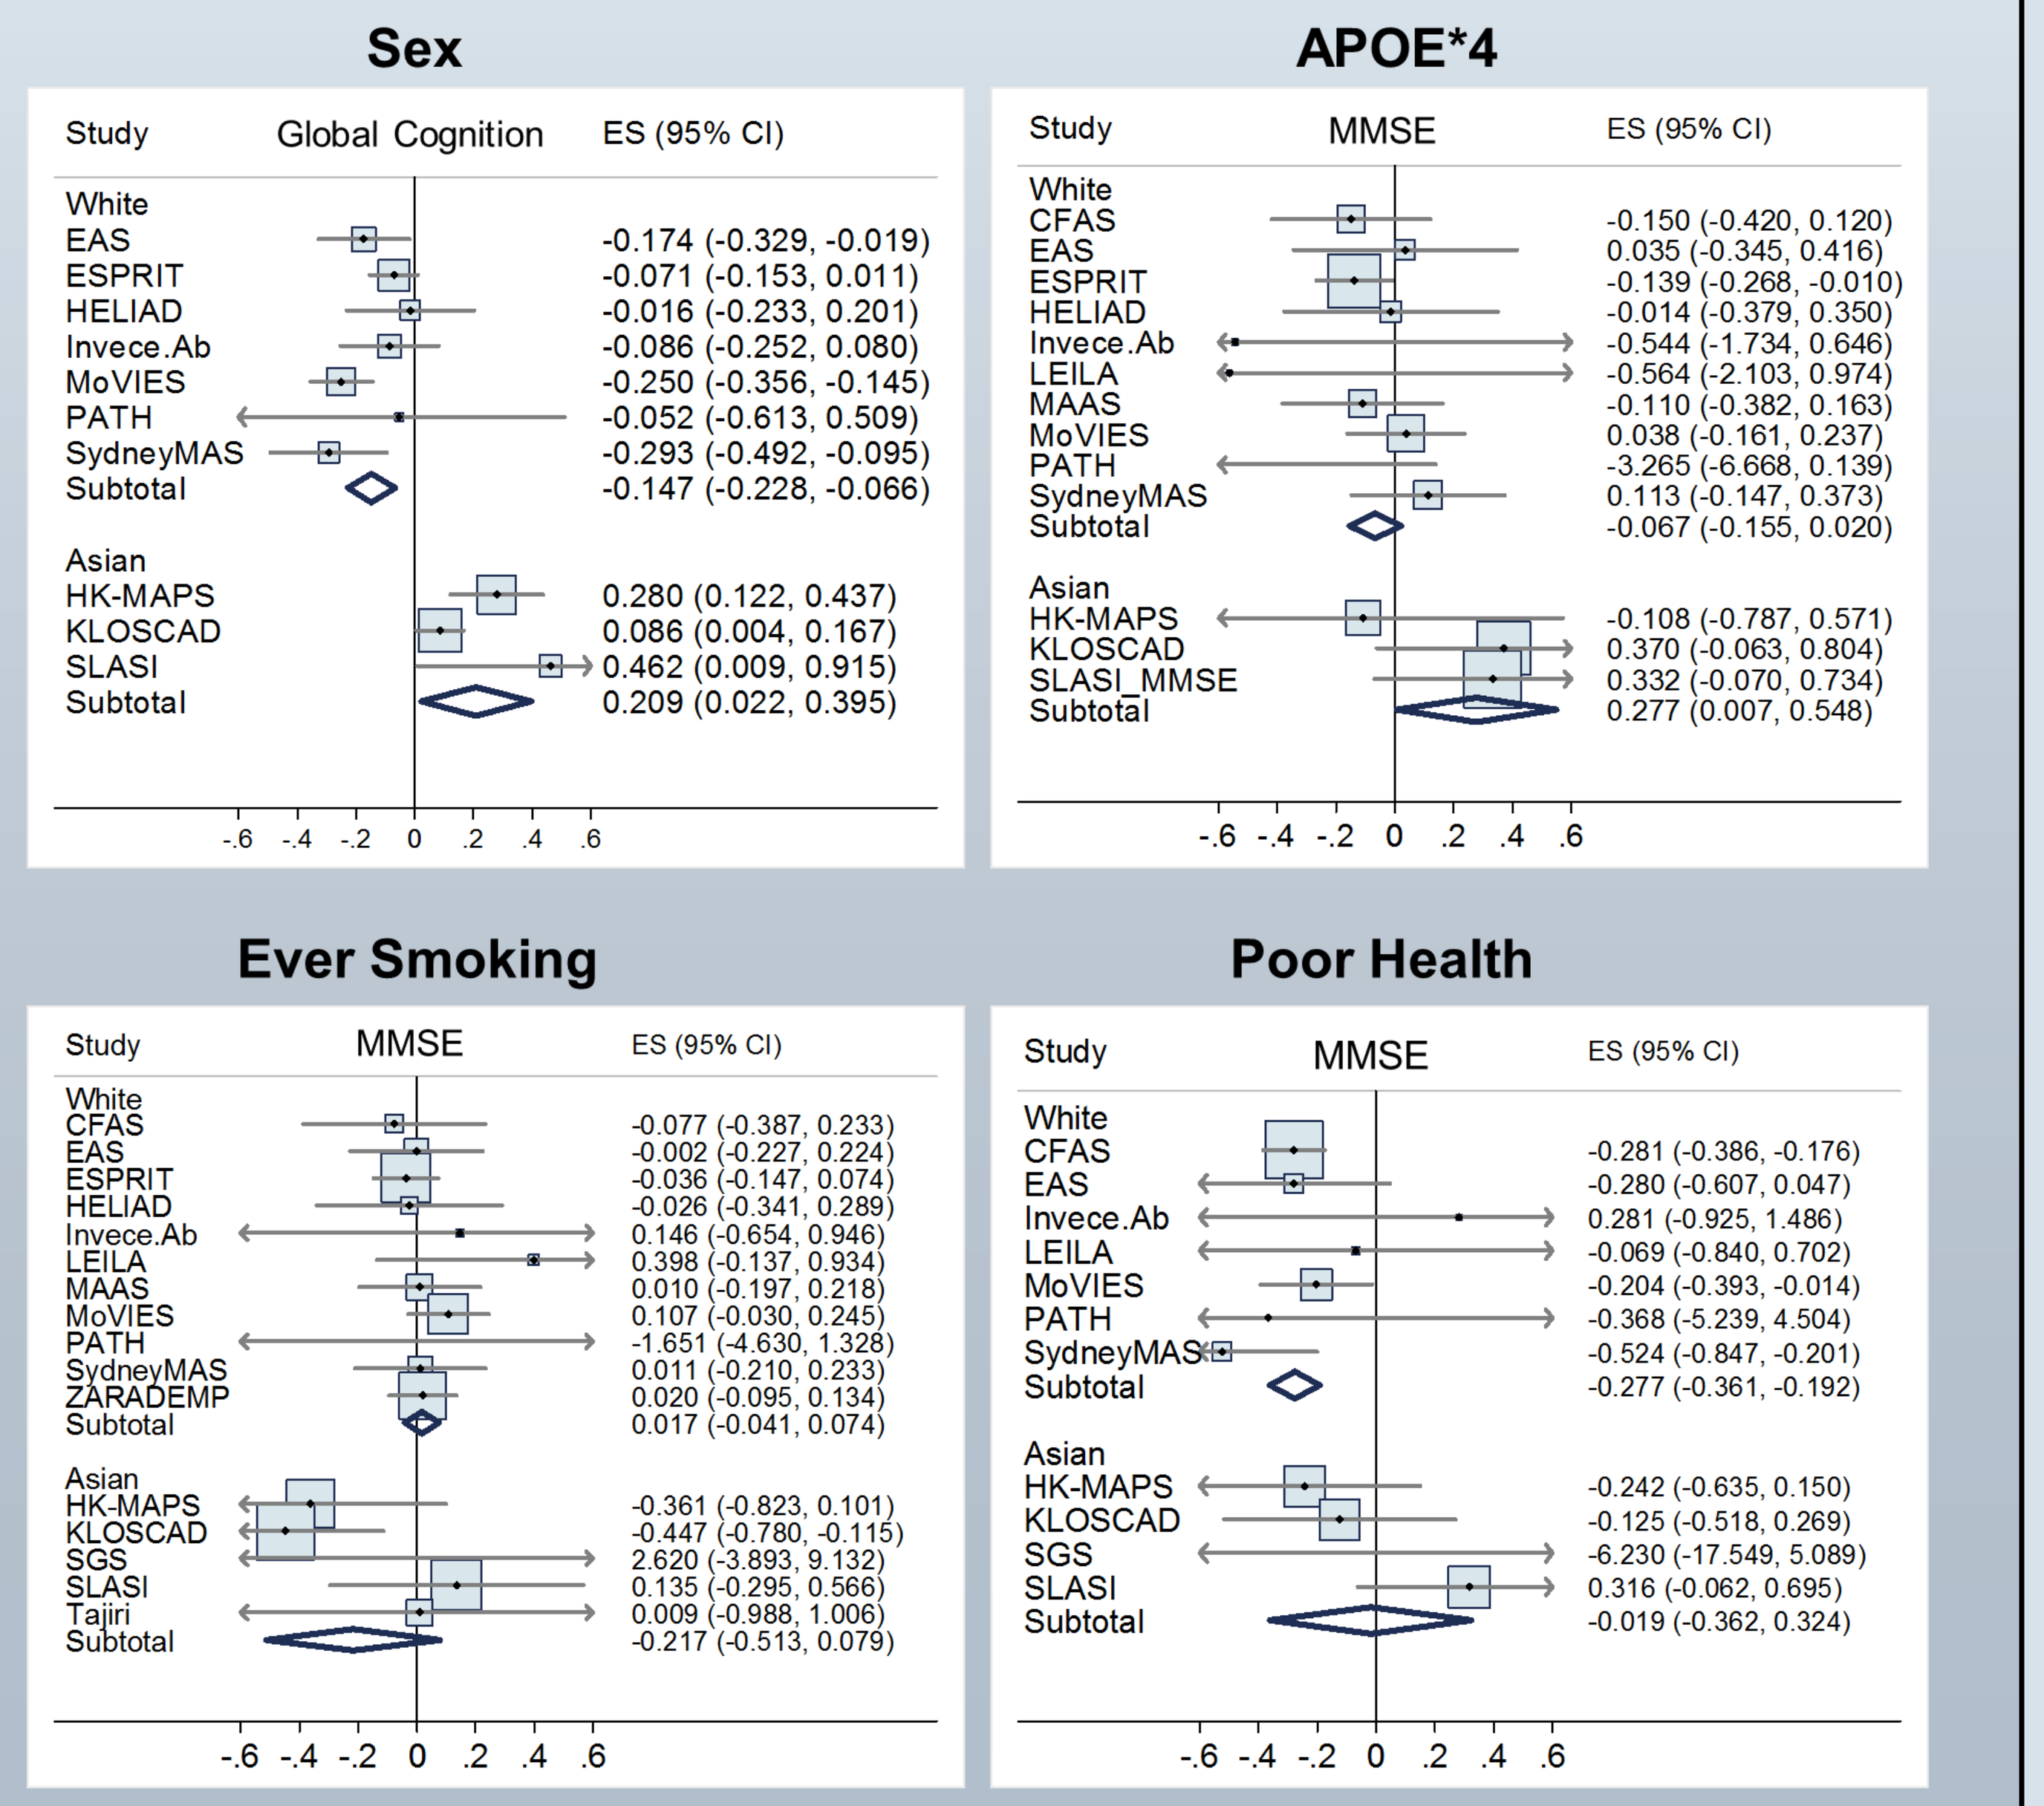
Sex
Study Global Cognition ES (95% CI)
White
EAS -0.174 (-0.329, -0.019)
ESPRIT -0.071 (-0.153, 0.011)
HELIAD -0.016 (-0.233, 0.201)
Invece.Ab -0.086 (-0.252, 0.080)
MoVIES -0.250 (-0.356, -0.145)
PATH -0.052 (-0.613, 0.509)
SydneyMAS -0.293 (-0.492, -0.095)
Subtotal -0.147 (-0.228, -0.066)
Asian
HK-MAPS 0.280 (0.122, 0.437)
KLOSCAD 0.086 (0.004, 0.167)
SLASI 0.462 (0.009, 0.915)
Subtotal 0.209 (0.022, 0.395)
APOE*4
Study MMSE ES (95% CI)
White
CFAS -0.150 (-0.420, 0.120)
EAS 0.035 (-0.345, 0.416)
ESPRIT -0.139 (-0.268, -0.010)
HELIAD -0.014 (-0.379, 0.350)
Invece.Ab -0.544 (-1.734, 0.646)
LEILA -0.564 (-2.103, 0.974)
MAAS -0.110 (-0.382, 0.163)
MoVIES 0.038 (-0.161, 0.237)
PATH -3.265 (-6.668, 0.139)
SydneyMAS 0.113 (-0.147, 0.373)
Subtotal -0.067 (-0.155, 0.020)
Asian
HK-MAPS -0.108 (-0.787, 0.571)
KLOSCAD 0.370 (-0.063, 0.804)
SLASI_MMSE 0.332 (-0.070, 0.734)
Subtotal 0.277 (0.007, 0.548)
Ever Smoking
Study MMSE ES (95% CI)
White
CFAS -0.077 (-0.387, 0.233)
EAS -0.002 (-0.227, 0.224)
ESPRIT -0.036 (-0.147, 0.074)
HELIAD -0.026 (-0.341, 0.289)
Invece.Ab 0.146 (-0.654, 0.946)
LEILA 0.398 (-0.137, 0.934)
MAAS 0.010 (-0.197, 0.218)
MoVIES 0.107 (-0.030, 0.245)
PATH -1.651 (-4.630, 1.328)
SydneyMAS 0.011 (-0.210, 0.233)
ZARADEMP 0.020 (-0.095, 0.134)
Subtotal 0.017 (-0.041, 0.074)
Asian
HK-MAPS -0.361 (-0.823, 0.101)
KLOSCAD -0.447 (-0.780, -0.115)
SGS 2.620 (-3.893, 9.132)
SLASI 0.135 (-0.295, 0.566)
Tajiri 0.009 (-0.988, 1.006)
Subtotal -0.217 (-0.513, 0.079)
Poor Health
Study MMSE ES (95% CI)
White
CFAS -0.281 (-0.386, -0.176)
EAS -0.280 (-0.607, 0.047)
Invece.Ab 0.281 (-0.925, 1.486)
LEILA -0.069 (-0.840, 0.702)
MoVIES -0.204 (-0.393, -0.014)
PATH -0.368 (-5.239, 4.504)
SydneyMAS -0.524 (-0.847, -0.201)
Subtotal -0.277 (-0.361, -0.192)
Asian
HK-MAPS -0.242 (-0.635, 0.150)
KLOSCAD -0.125 (-0.518, 0.269)
SGS -6.230 (-17.549, 5.089)
SLASI 0.316 (-0.062, 0.695)
Subtotal -0.019 (-0.362, 0.324)
-.6 -.4 -.2 0 .2 .4 .6

Fig 1. Forest plots displaying regression coefficients for associations between individual factors and cognitive performance in studies comprised primarily of white or Asian participants.

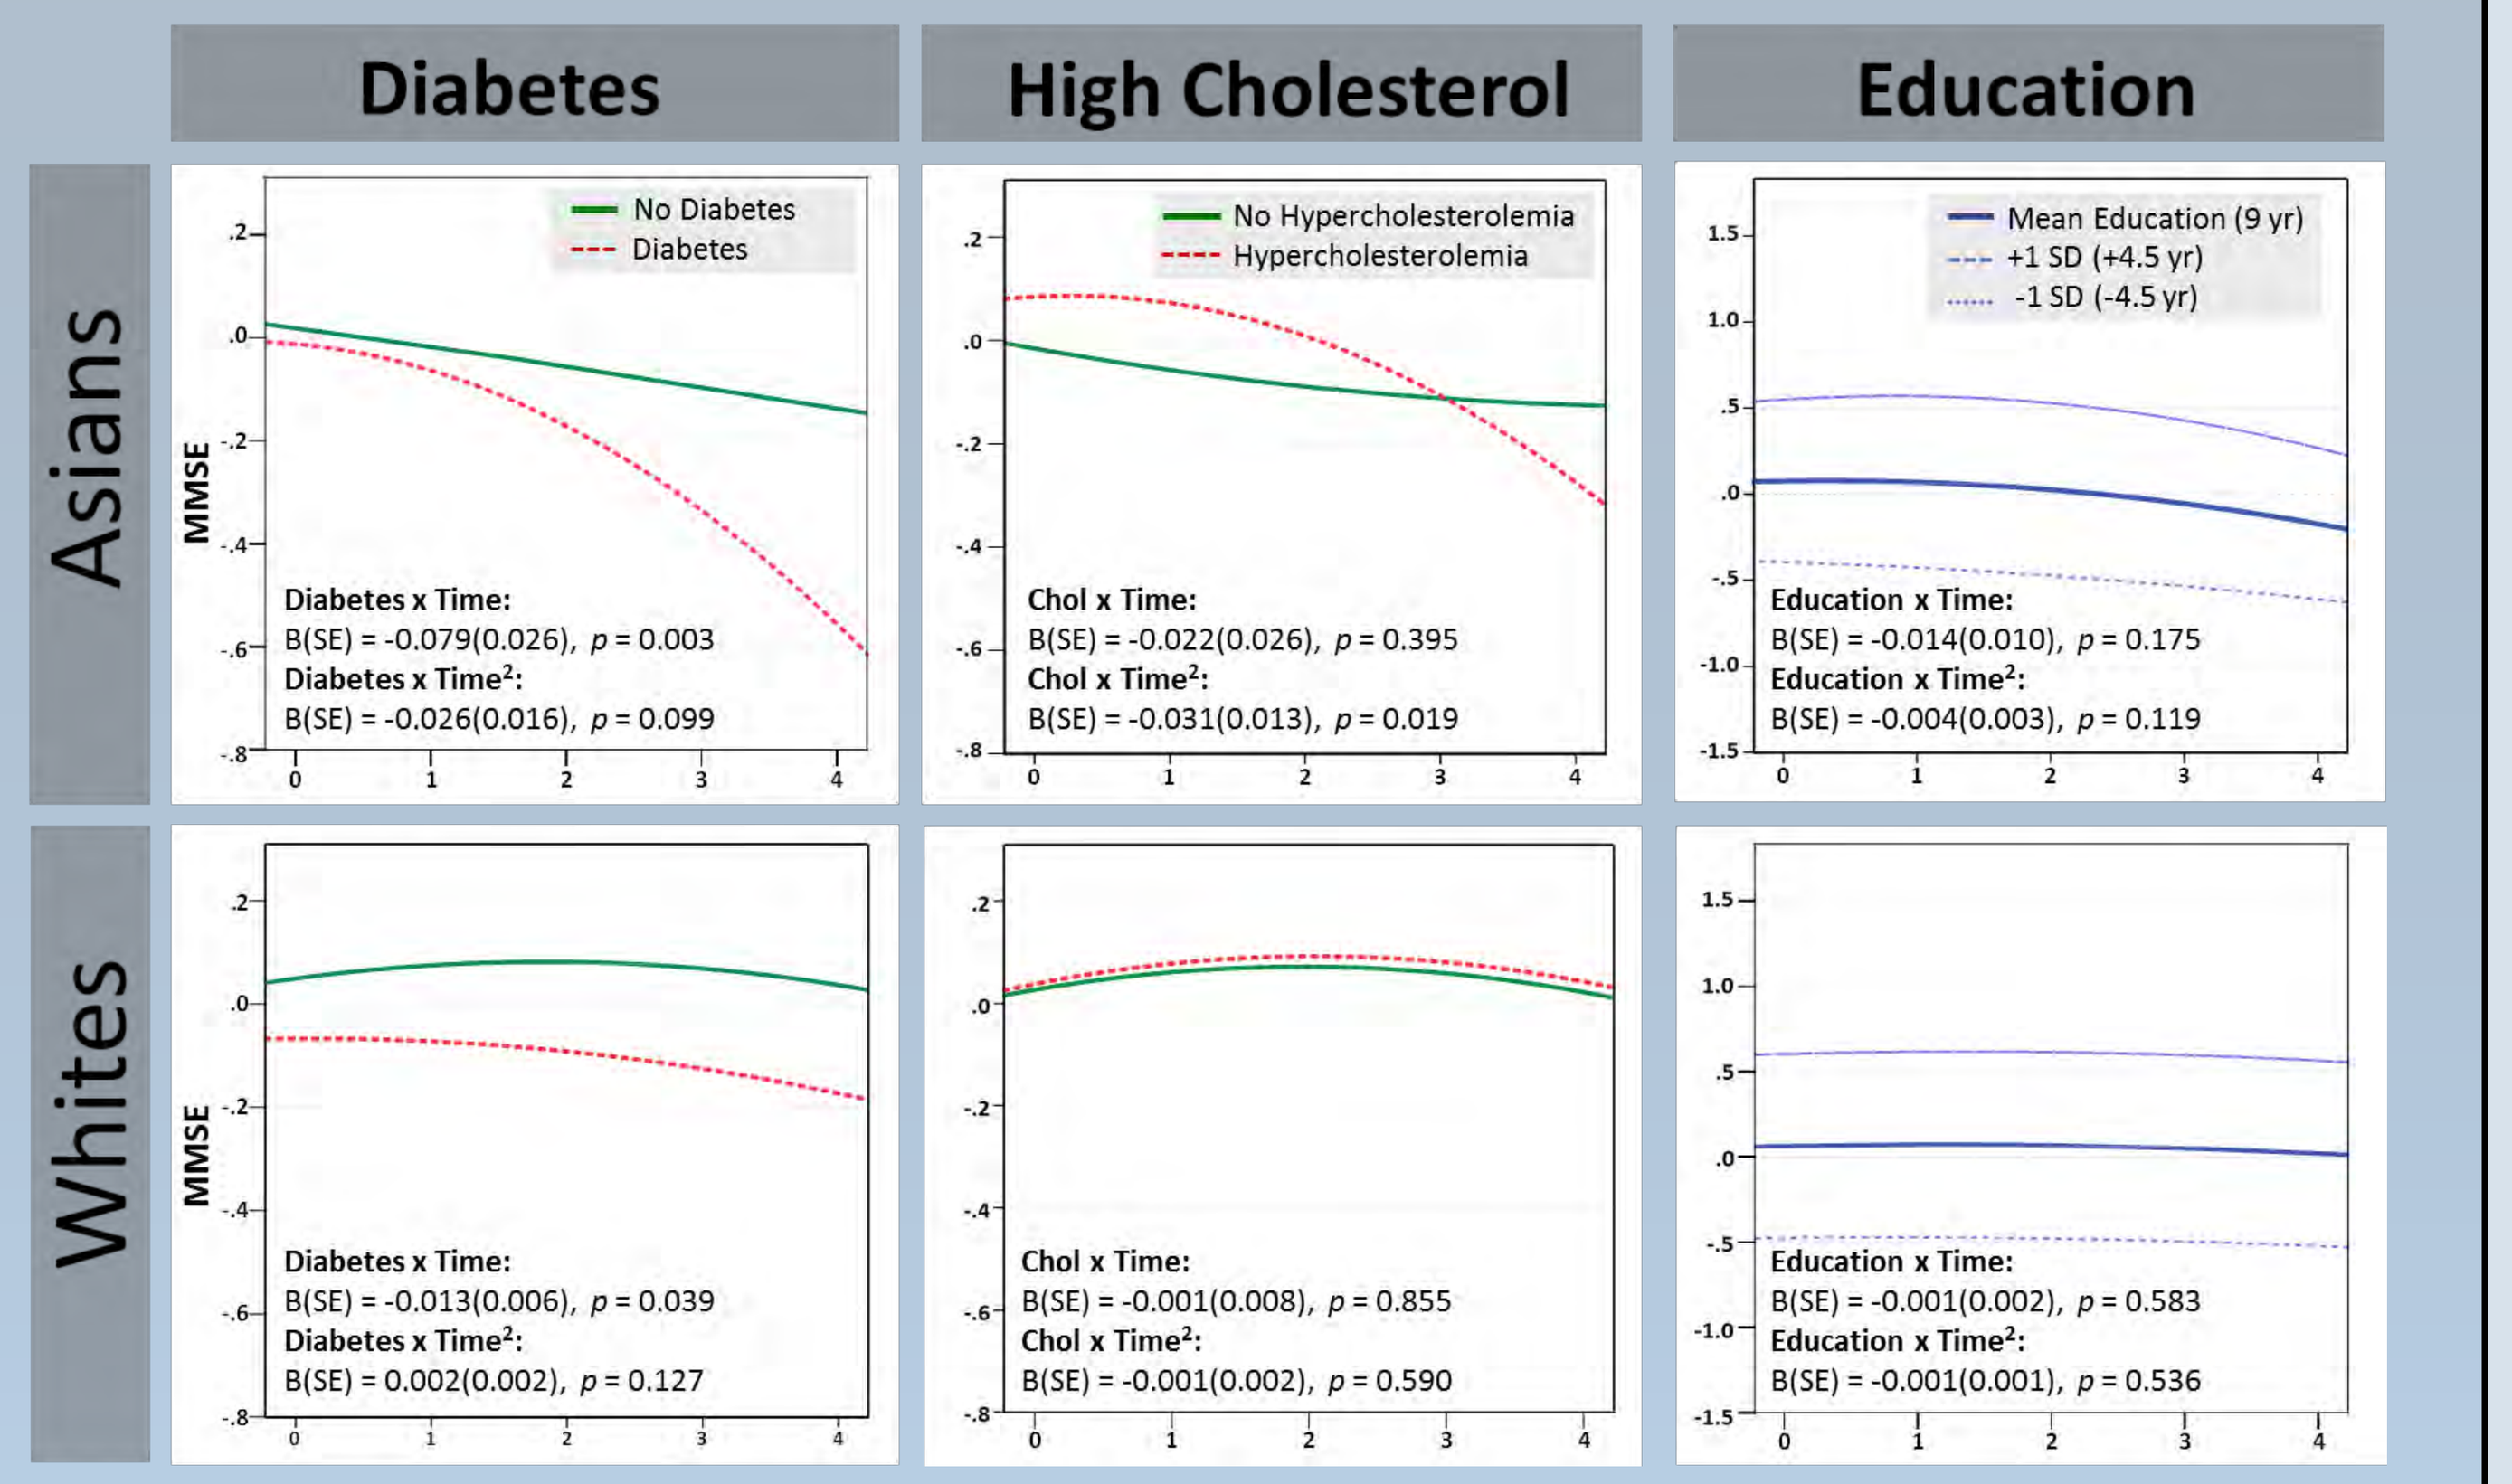
Diabetes
High Cholesterol
Education
Asians
Whites
MMSE
No Diabetes
Diabetes
Diabetes x Time:
B(SE) = -0.079(0.026), p = 0.003
Diabetes x Time²:
B(SE) = -0.026(0.016), p = 0.099
No Hypercholesterolemia
Hypercholesterolemia
Chol x Time:
B(SE) = -0.022(0.026), p = 0.395
Chol x Time²:
B(SE) = -0.031(0.013), p = 0.019
Mean Education (9 yr)
+1 SD (+4.5 yr)
-1 SD (-4.5 yr)
Education x Time:
B(SE) = -0.014(0.010), p = 0.175
Education x Time²:
B(SE) = -0.004(0.003), p = 0.119
Diabetes x Time:
B(SE) = -0.013(0.006), p = 0.039
Diabetes x Time²:
B(SE) = 0.002(0.002), p = 0.127
Chol x Time:
B(SE) = -0.001(0.008), p = 0.855
Chol x Time²:
B(SE) = -0.001(0.002), p = 0.590
Education x Time:
B(SE) = -0.001(0.002), p = 0.583
Education x Time²:
B(SE) = -0.001(0.001), p = 0.536

Fig 2. Fitted trajectories for whites and Asians comparing changes in MMSE scores (standardized values on the Y-axis) over time in years (X-axis) for factors showing associations with cognitive decline or rate of change in decline that differed between the groups.

Conclusions

We considered the most important factors those that combined factor models identified as independently associated with cognitive performance or decline.

- Ageing and being *APOE*4* positive have no direct resolution (currently), and it is not clear whether depression is a cause or prodrome of decline.

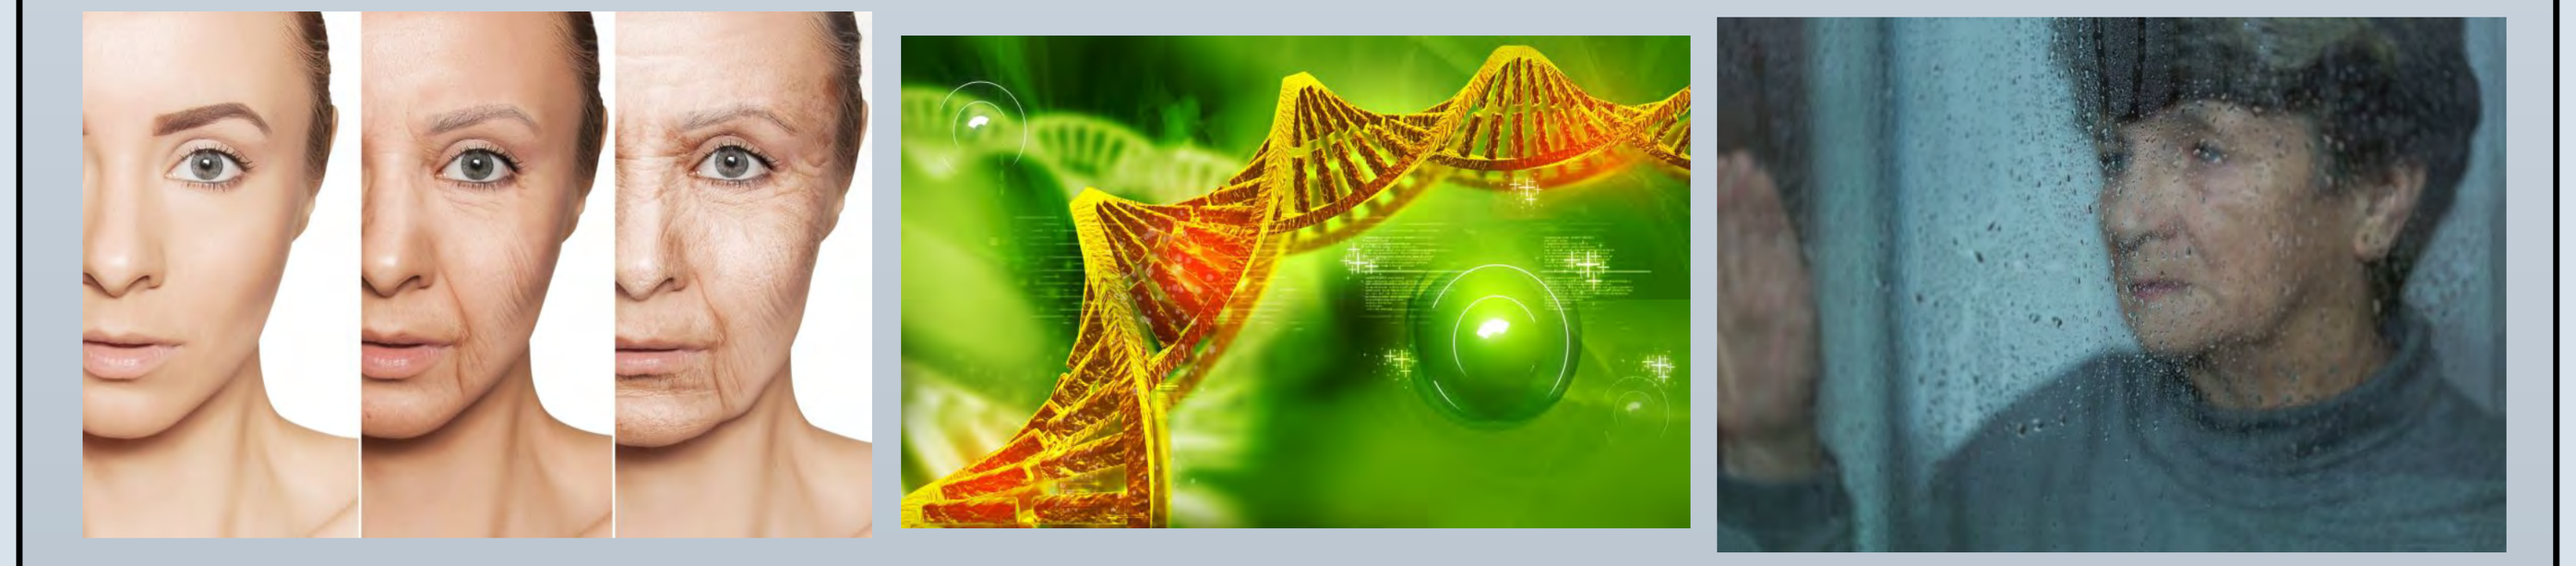

- Other factors are modifiable, suggesting or strengthening support for targeting them in interventions to delay or minimize cognitive decline, including that leading to dementia:
 - Increased levels of education
 - Being physically active
 - Not smoking
 - Controlling other factors that lower the risk of diabetes and stroke.

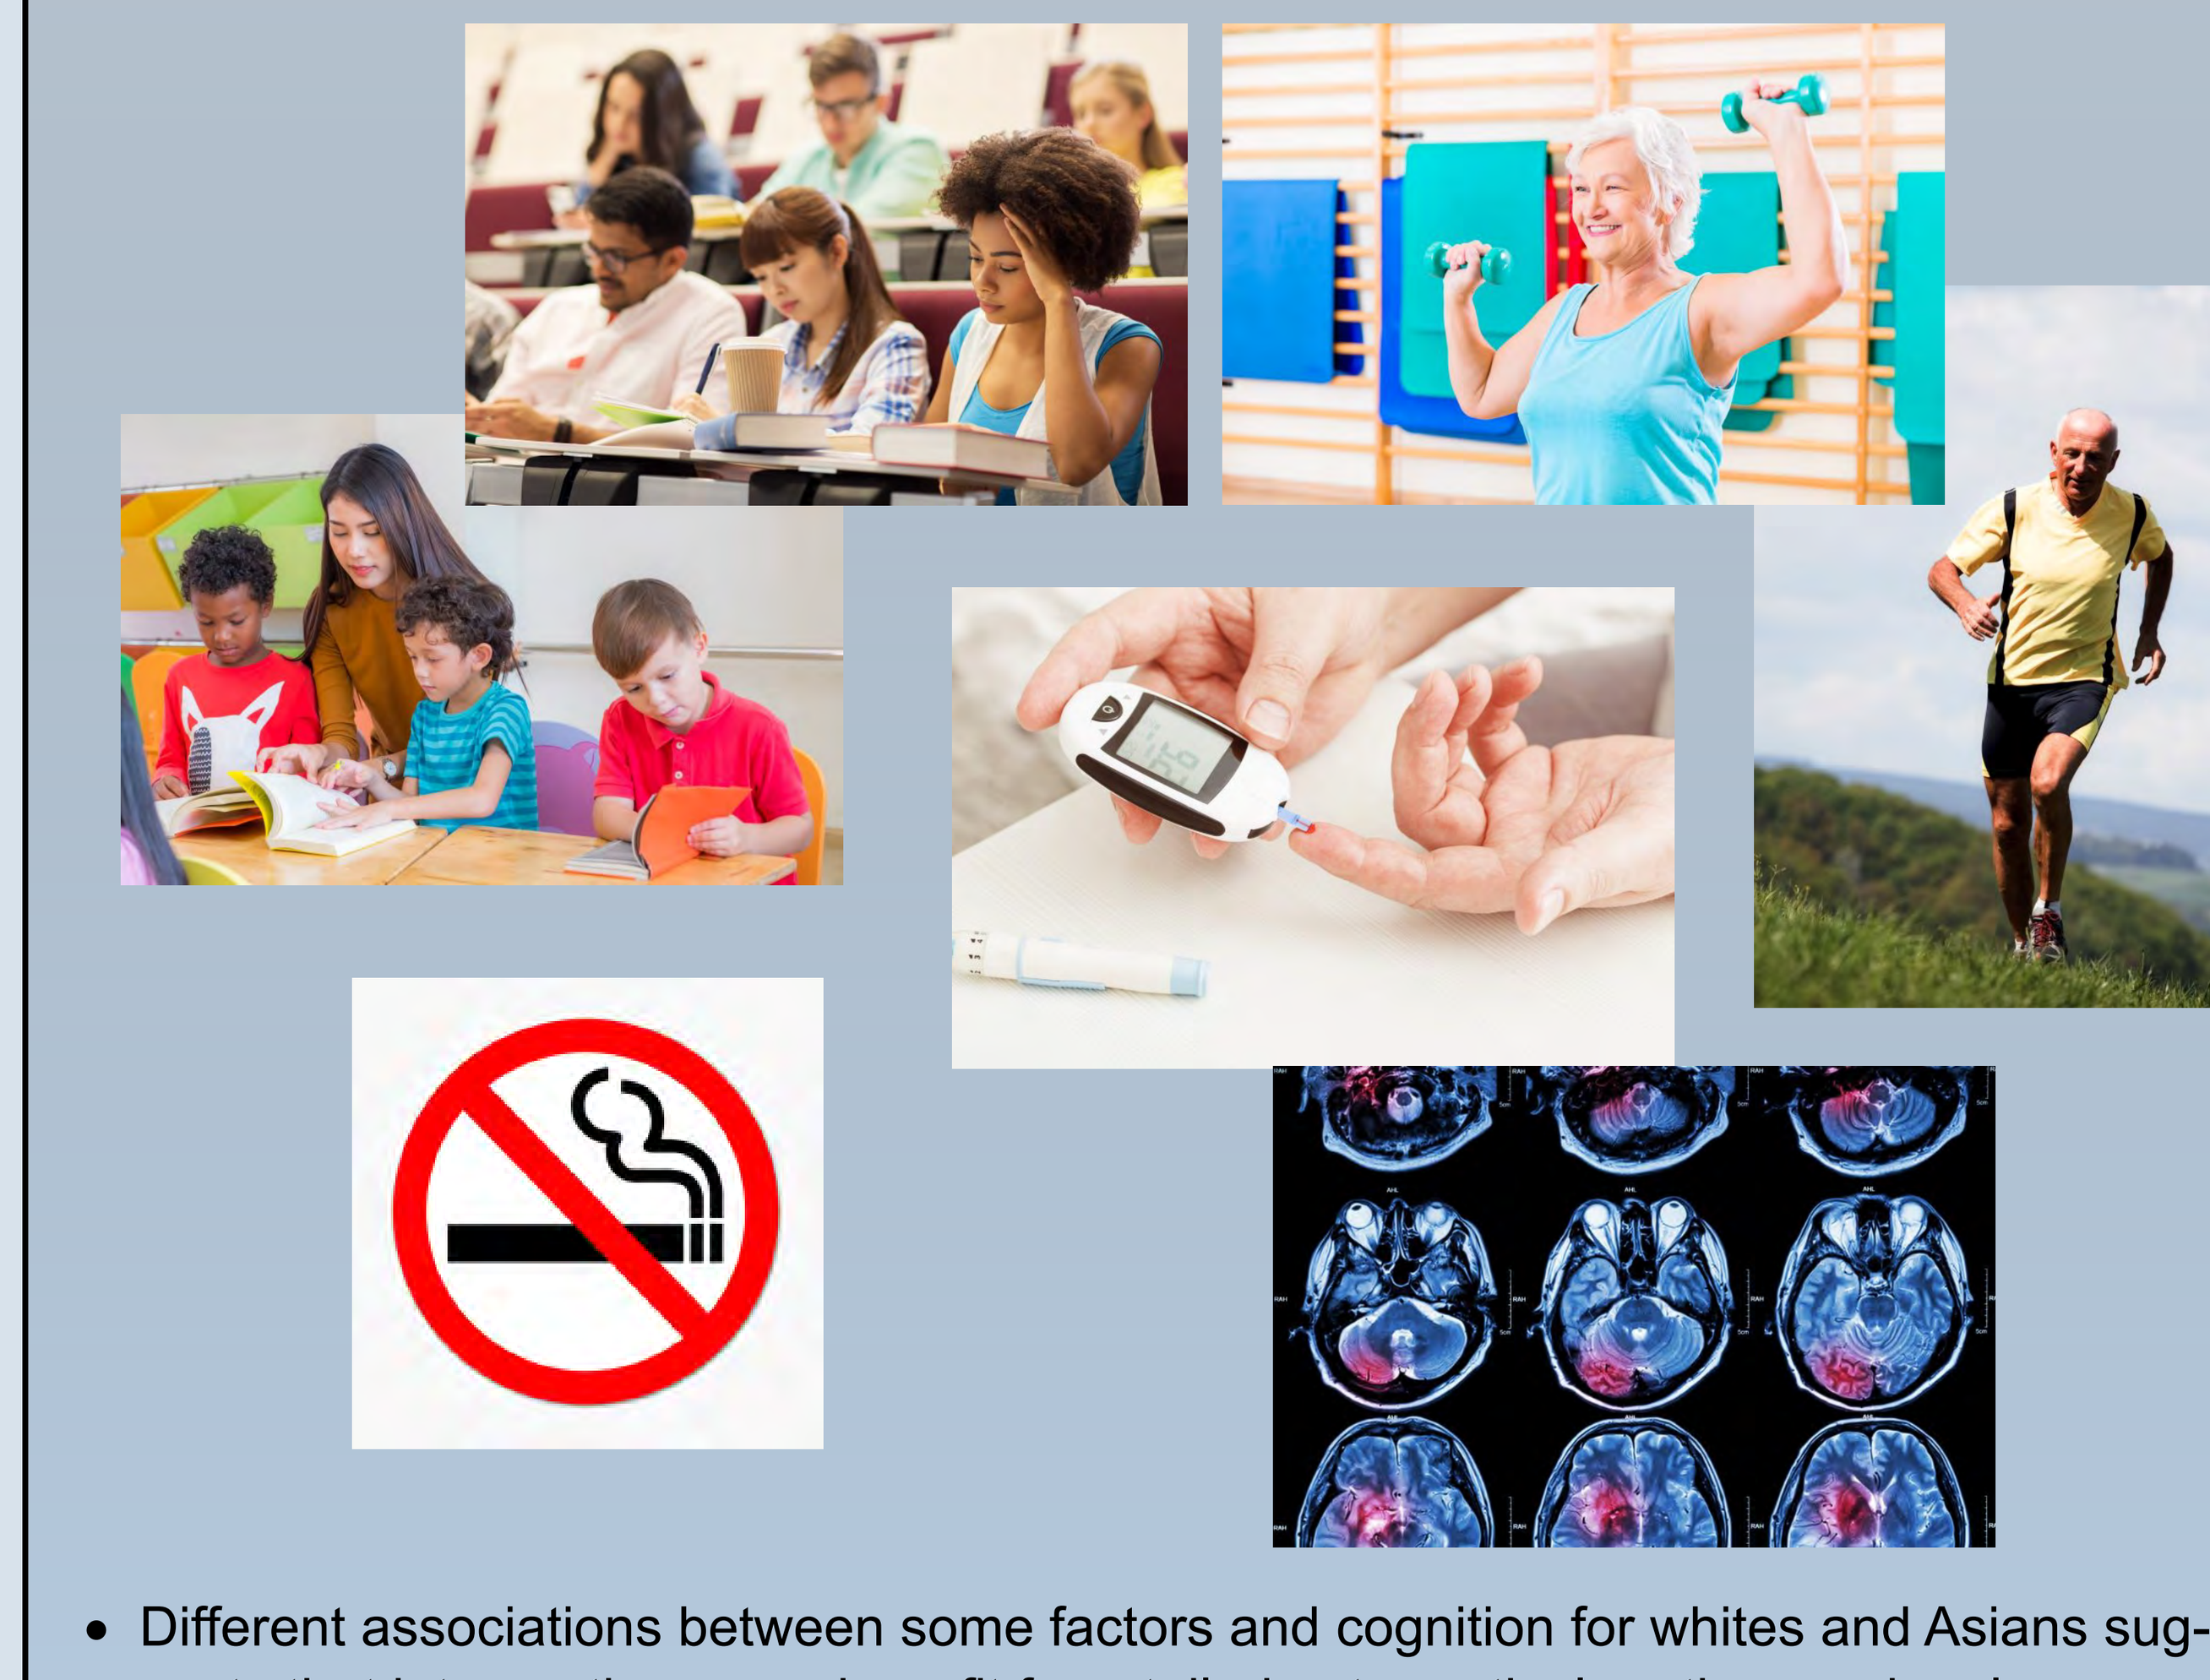

- Different associations between some factors and cognition for whites and Asians suggests that interventions may benefit from tailoring to particular ethno-regional groups.

Contact

All researchers can apply to use COSMIC data. For details please contact:
Darren Lipnicki (COSMIC Study Co-ordinator), d.lipnicki@unsw.edu.au
Perminder Sachdev (COSMIC head), p.sachdev@unsw.edu.au
Website: https://cheba.unsw.edu.au/group/cosmic